1243

A Patient-focused Program for Using Steroids Wisely

Authors: John Stone, MD, MPH, Michelle Petri, MD, MPH, Jeffrey Gelfand, MD, Camille Kotton, MD, Jane McDowell, MBBS, BSc, PhD, George Papaliodis, MD, Meredith Marinaro, PhD, RD, Matt Wilkinson, PhD, MBA, Walter Lentfert, BS, Martha Stone, BA

Equipping Patients for Shared Decision-making

Abstract

Background/Purpose

Patients have no comprehensive, curated resource on steroid use to facilitate effective collaboration in their own care. Consequently, truly shared decision-making around steroid treatment is challenging. The Sam program - short for “Steroids and Me” - is a collaboration among academic physicians, patients, patient advocacy groups and industry leaders to develop a patient-focused program to help patients optimize their steroid use. We report on Sam’s two years of development and first five months of use.

Methods

Sam’s development plan was divided into three phases. Phase 1 (Resource Review) was a survey of the medical literature and internet for current, scientifically rigorous, patient-focused content on steroid use in clinical care. Phase 2 (Needs Assessment) consisted of two international roundtable meetings of patient advocacy group leaders and industry representatives (total participants, n=36); separate meetings with 6 patient advocacy groups; three patient focus groups; and meetings with industry representatives from five companies. Finally, Phase three (Sam Build) consisted of building the web-based platform accessible by phone, tablet or computer. In this phase, a 34-member patient group was engaged in one-on-one meetings for user testing and patient journey validation.

Results

Sam is protected by stringent privacy policies and fulfills requirements of both HIPAA/GDPR and state-specific data privacy laws. The development program launched at ACR Convergence 2022 and was researched and built over a two-year period. In Phase 1 (Resource Review), audits were performed on 54 patient advocacy and patient-facing industry websites. Although the medical literature contains abundant documentation of the damage caused by steroid use, there is no dedicated instruction for patients about how to mitigate these side-effects: the Resource Review identified not a single source of curated, comprehensive, patient-focused material on steroid-toxicity. In Phase 2 (Needs Assessment), patient advocacy groups representing lupus, vasculitis, pemphigus, myasthenia gravis, sarcoidosis and IgG4-related disease participated. Discussions with individual patients and groups of patients identified major unmet needs that were amplified by patient advocacy group leaders and industry.

All Sam visitors can access educational content, including written material on dozens of known steroid side effects; tips about the prevention and management of steroid-toxicities; and videos created by physician experts to supplement those educational tools.
All content is original, developed and recorded by internationally-recognized clinicians. Sam also provides tools, including mood and side effect tracking, that prepare patients for shared decision-making conversations with their providers. Sam collects and stores longitudinal patient-reported data and displays a 30-day look back to summarize side effects in anticipation of follow-up clinic visits.

Conclusion

Steroids and Me (Sam) is a unique, patient-focused resource that engages, educates and equips users to understand and reduce steroid-toxicity, facilitating conversations that foster shared decision-making around steroid use.

Engagement

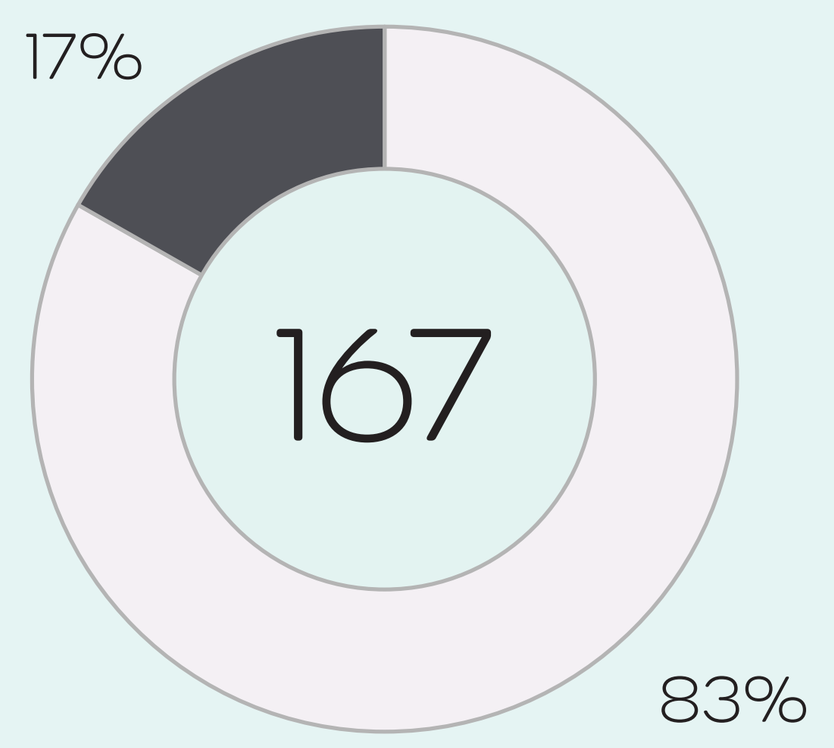

17%
167
83%

Visitors and registered users

Of the 167 patients from the Vasculitis Foundation interacting with the Sam website, 28 (17%) registered as Sam users, providing their age, ethnicity, condition, time on steroids and sex.

Visitors have full access to education and registered users have full access to education and tracking tools.

Session time in minutes

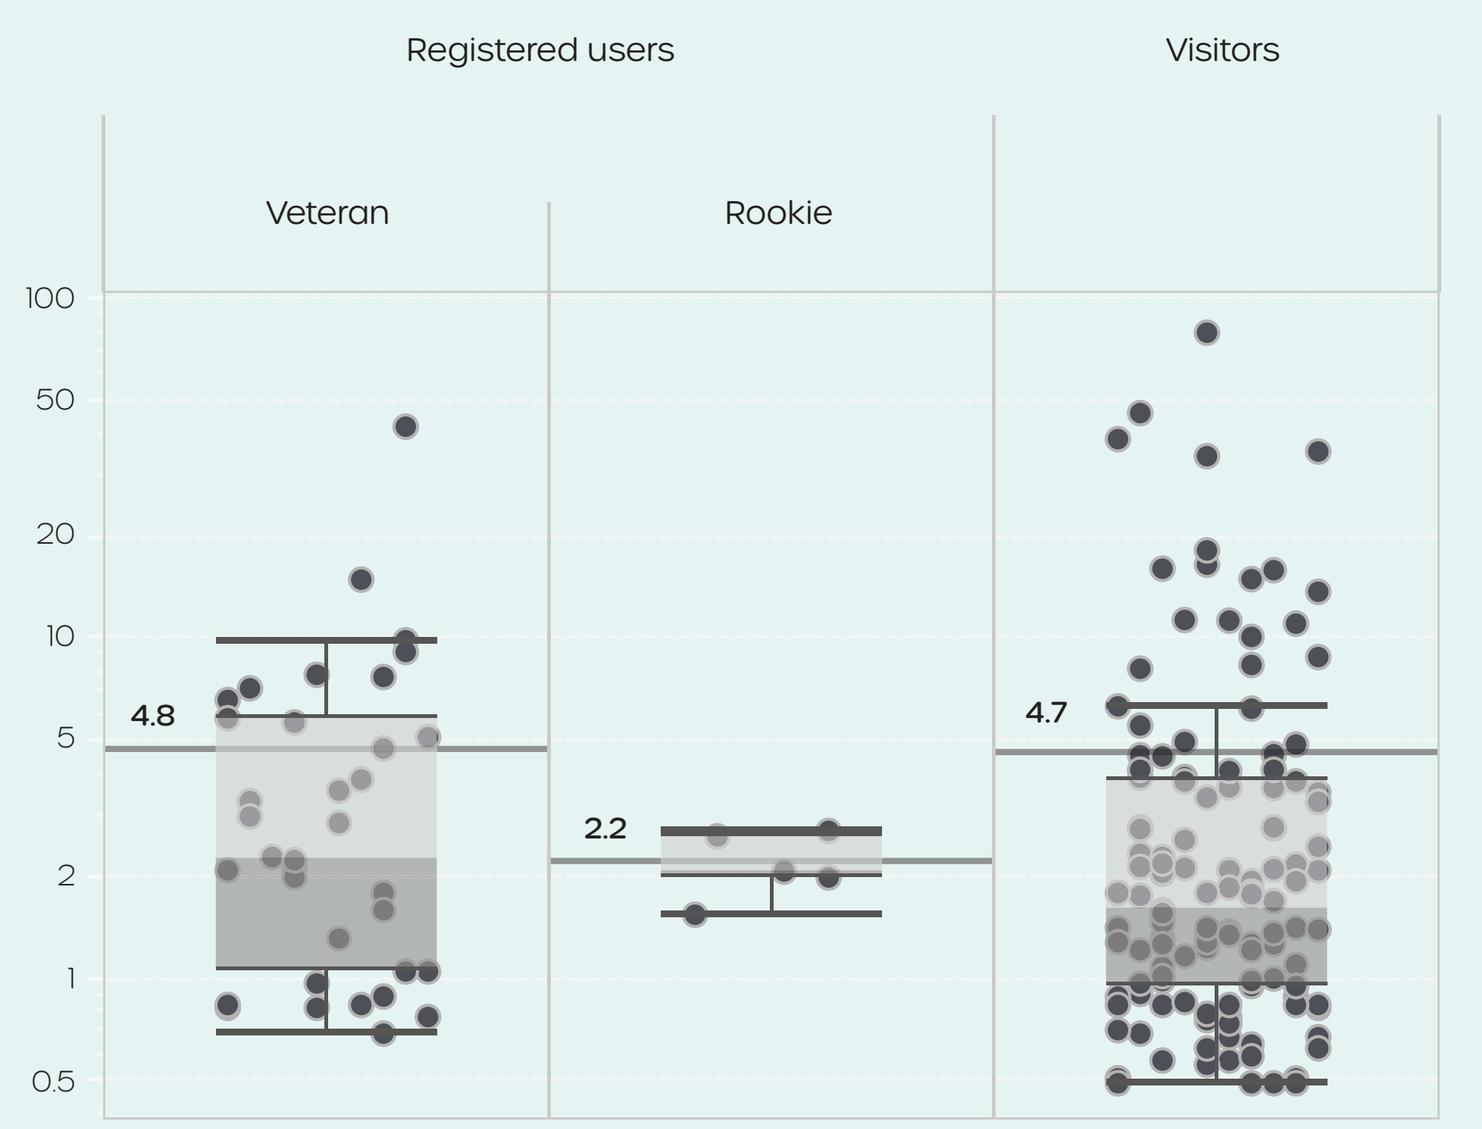

Registered users
Visitors
Veteran
Rookie
100
50
20
10
5
2
1
0.5
4.8
2.2
4.7

Engagement

	Registered users (28)	Visitors (139)
Page views	265	543
Pages per user	9	4
Sessions	42	133
Sessions per user*	1.5	1.0

* returning visitors are not tracked, so subsequent sessions for the same visitor will be tracked as a new user.

Visitors and registered users

	Registered users (28)		Visitors (139)	
Welcome	97	37%	303	56%
Exploring side effects	56	21%	114	21%
Reading articles	29	11%	126	23%
Patient journey	83	31%	N/A*	
Grand total	265	100%	543	100%

* Patient journey is available to registered users only.

Key findings

A single invitation to Vascultitis Foundation members resulted in:

- **Heavy engagement in learning about steroid-toxicity**
- **High conversion rate: 18% of all visitors became registered users**
- **Major interest in “Why we taper” and “How to talk to my doctor”**
- **Average number of articles read: 5**
- **Average number of side effects tracked: 4**
- **Average session time: 5 minutes with a maximum of 100 minutes**

Learning

Top 5 topics read

Why we taper	29
Talking to my doctor	25
Weight and appearance	18
Sleep	16
Mental health	14

Of the 15 articles in Sam, these five topics collectively accounted for 102 separate content engagements, or 66% of all content engagements.

Number of articles read by all: Registered users and visitors

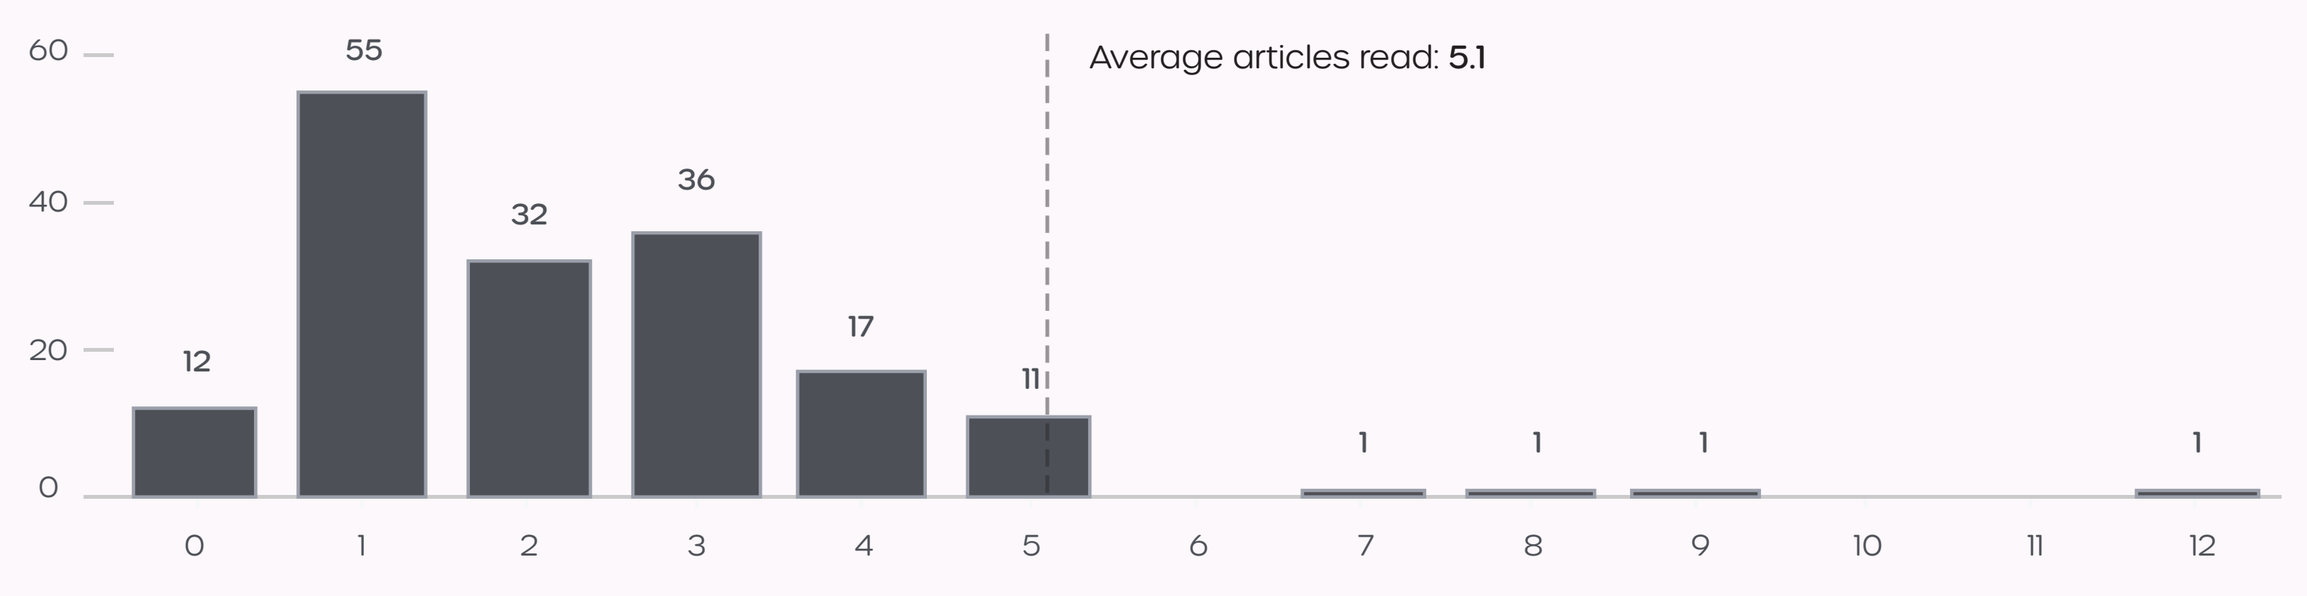

Average articles read: 5.1
60
40
20
0
12
55
32
36
17
11
1
1
1
1
0 1 2 3 4 5 6 7 8 9 10 11 12

Top 5 side effects recorded

Fatigue	19
Skin	14
Weight and appearance	16
Mental health	12
Bone health	9

The top 5 side effects recorded by Registered Users reflect common and noticeable signs of steroid toxicity.

Number of side effects recorded per registered user

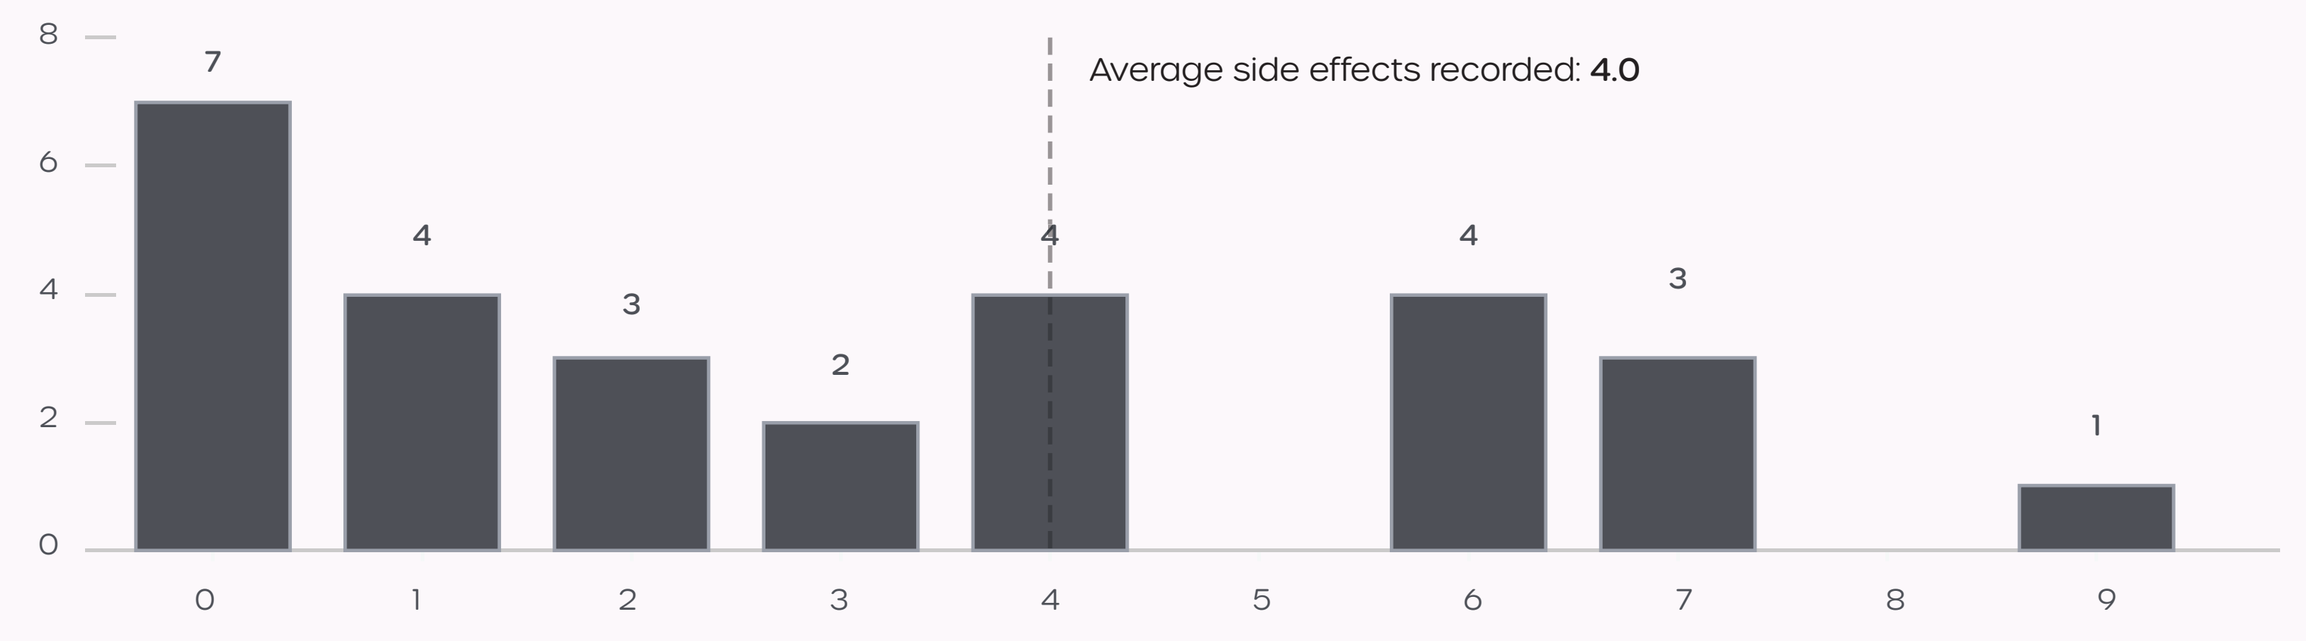

Average side effects recorded: 4.0
8
6
4
2
0
7
4
3
2
4
4
3
1
0 1 2 3 4 5 6 7 8 9

Demographics

Steroid experience

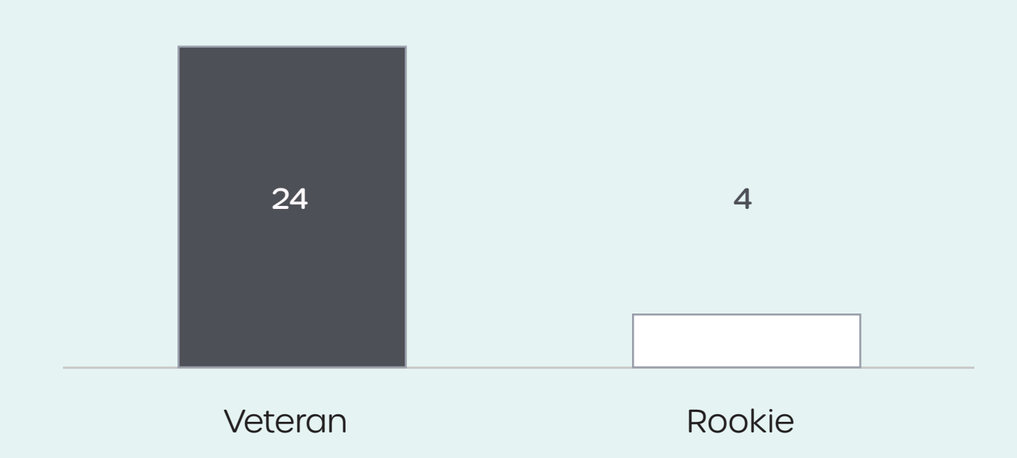

24
4
Veteran
Rookie

Age distribution

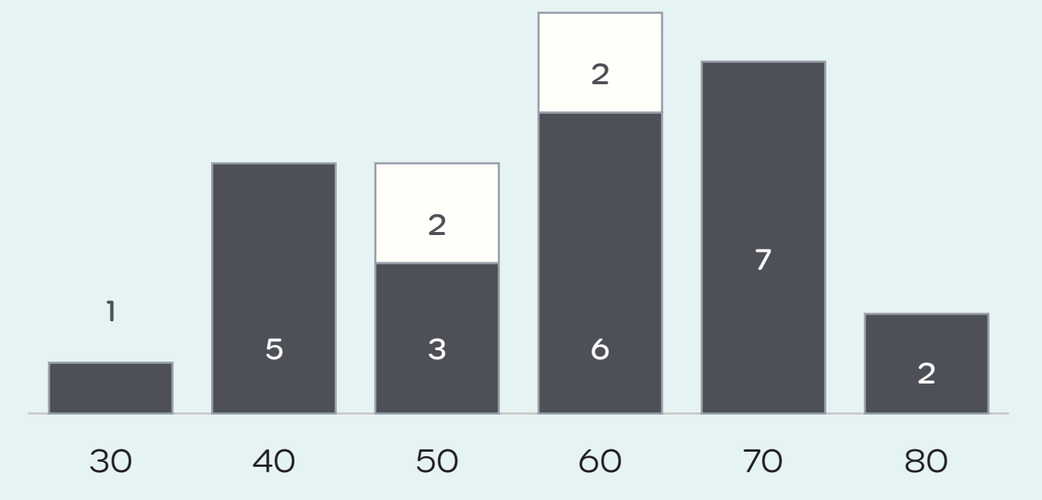

1
5
2
3
2
6
7
2
30 40 50 60 70 80

Ethnicity

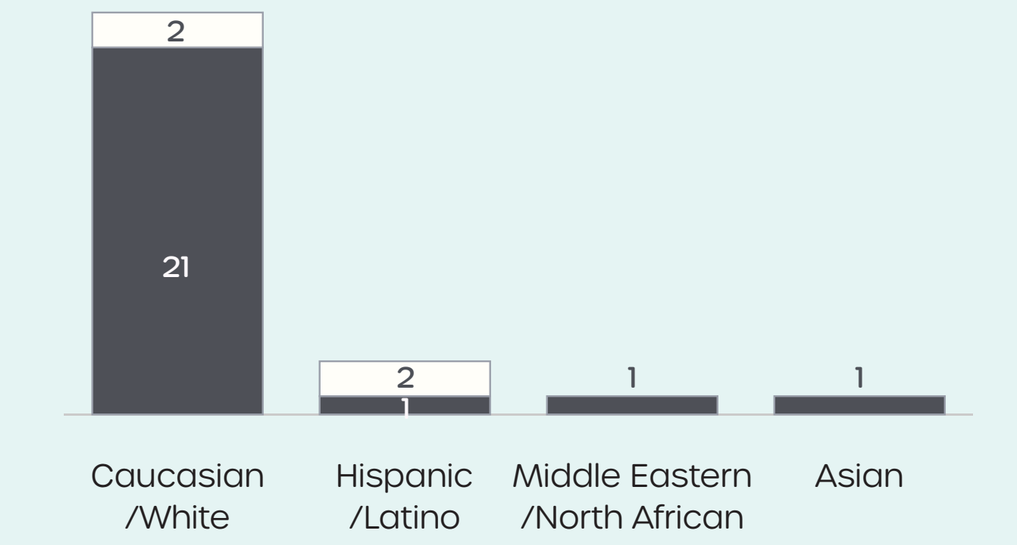

2
21
2
1
1
1
Caucasian /White
Hispanic /Latino
Middle Eastern /North African
Asian

Sex

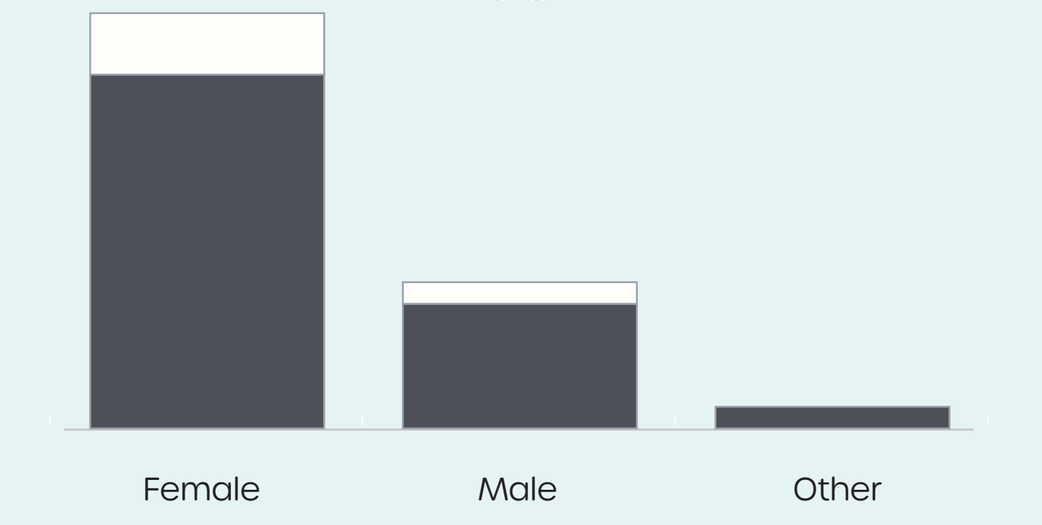

Female
Male
Other

Key: Veteran — Registered user reporting > 6 months total steroid use; Rookie — Registered user reporting < 6 months total steroid use

Diagnosis

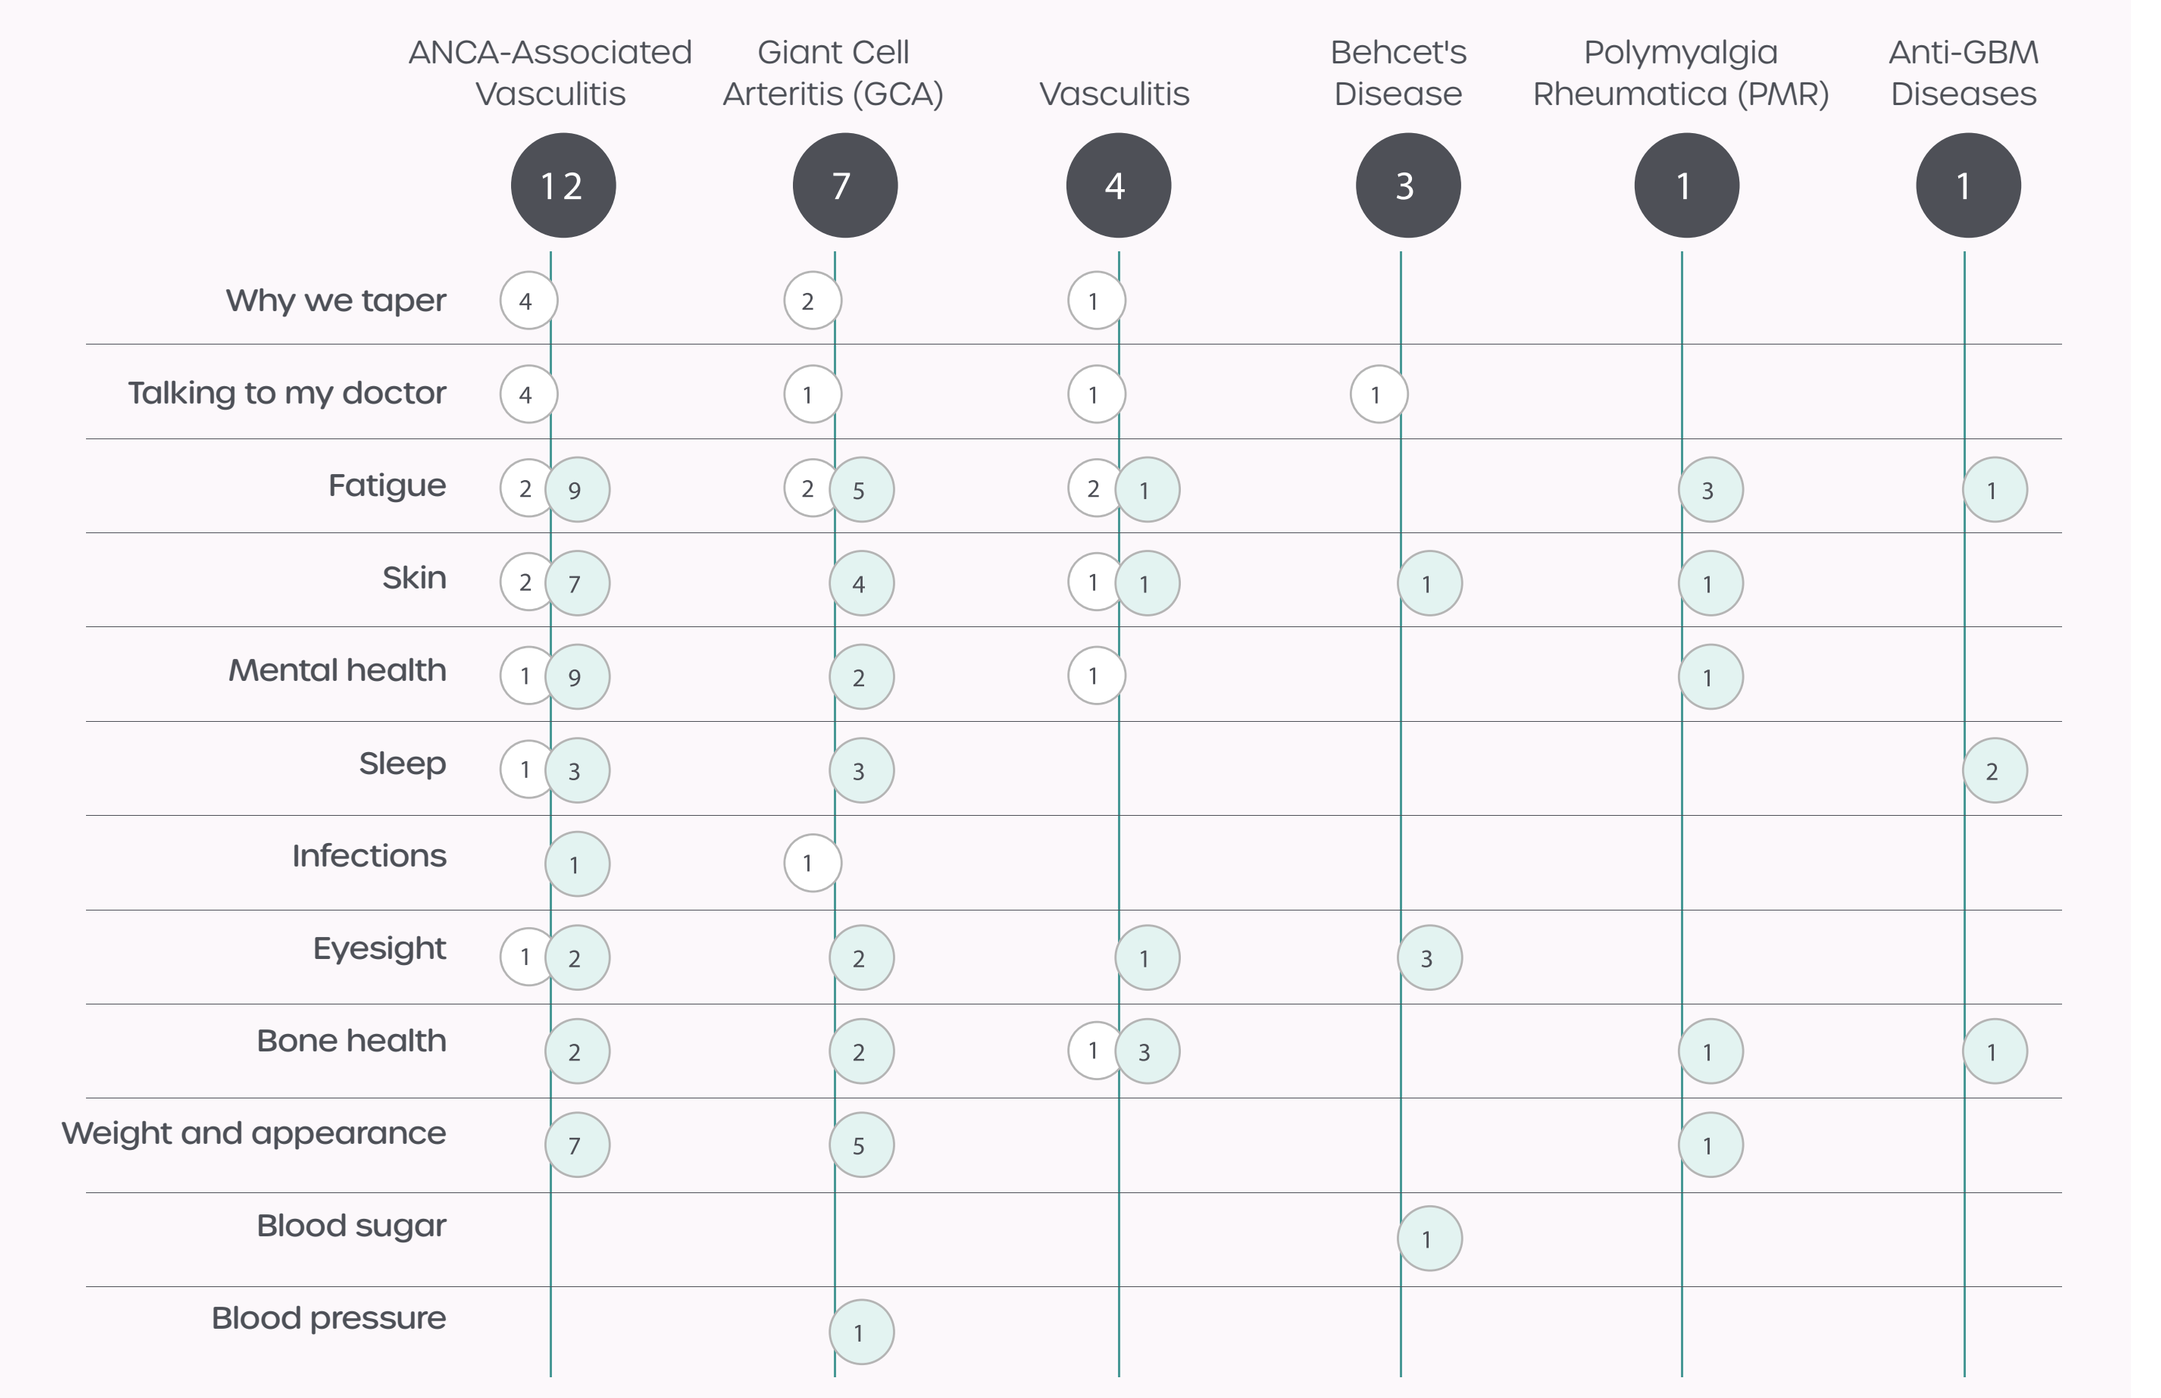

ANCA-Associated Vasculitis
Giant Cell Arteritis (GCA)
Vasculitis
Behcet's Disease
Polymyalgia Rheumatica (PMR)
Anti-GBM Diseases
12
7
4
3
1
1
Why we taper
Talking to my doctor
Fatigue
Skin
Mental health
Sleep
Infections
Eyesight
Bone health
Weight and appearance
Blood sugar
Blood pressure

Key: Side effects recorded; Articles read

Development suppport from abbvie, Amgen, argenx, Steritas, Zenas

Steroid-treated Patients Voiced Unmet Need

- Patients voiced feelings of isolation in their steroid journey, noting the overwhelming and often contradictory information about steroids on the internet.
- They observed a lack of adequate resources to educate them about the optimal way to use steroids and how to talk with their doctors about steroid-sparing approaches.
- Patients commented that educating themselves about steroids while they are also learning about their underlying disease is daunting.
- Their search for information is complicated by the neuropsychiatric effects of steroids themselves (insomnia, depression, anxiety and altered cognition).
- The patients requested a trusted source where all relevant material with the potential to inform their journey was collected in one place.
- Patients recognized the value of original, contemporary and curated content from well-known/trusted clinicians, investigators and experts in inflammatory disease.
- Patients noted that although medical literature includes many of the bad effects of steroids, there is little information about what patients and their healthcare providers can do to mitigate them.

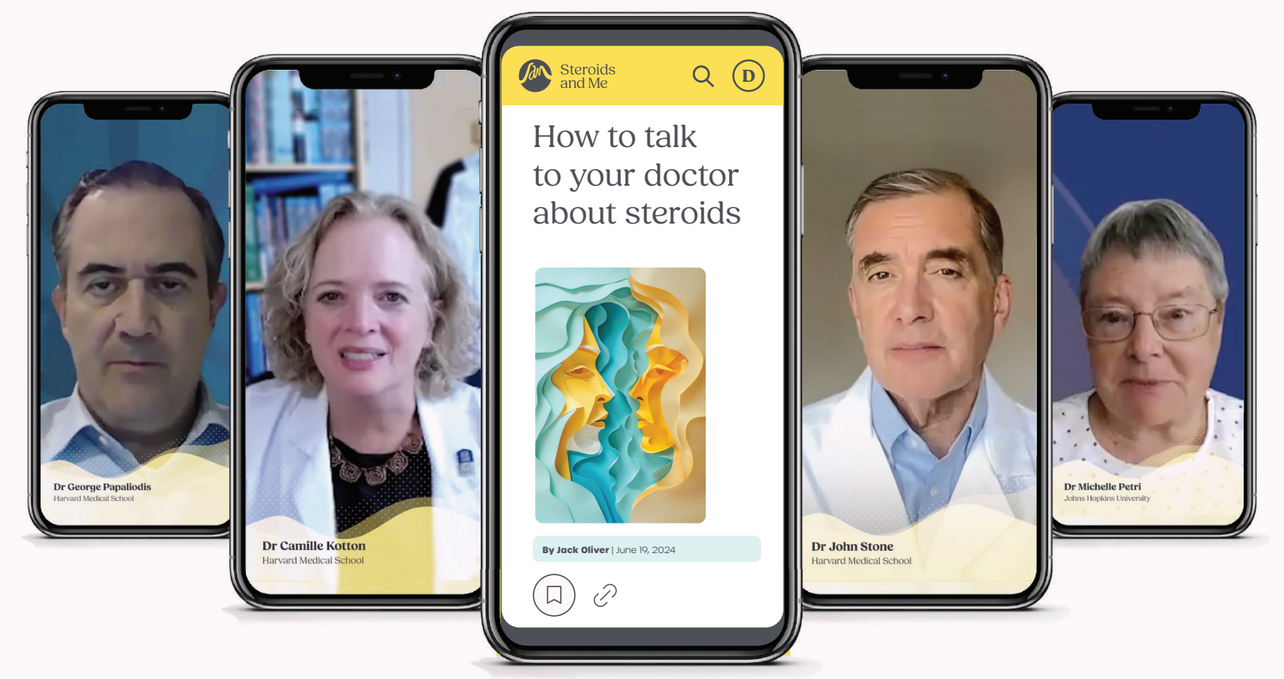

How to talk to your doctor about steroids

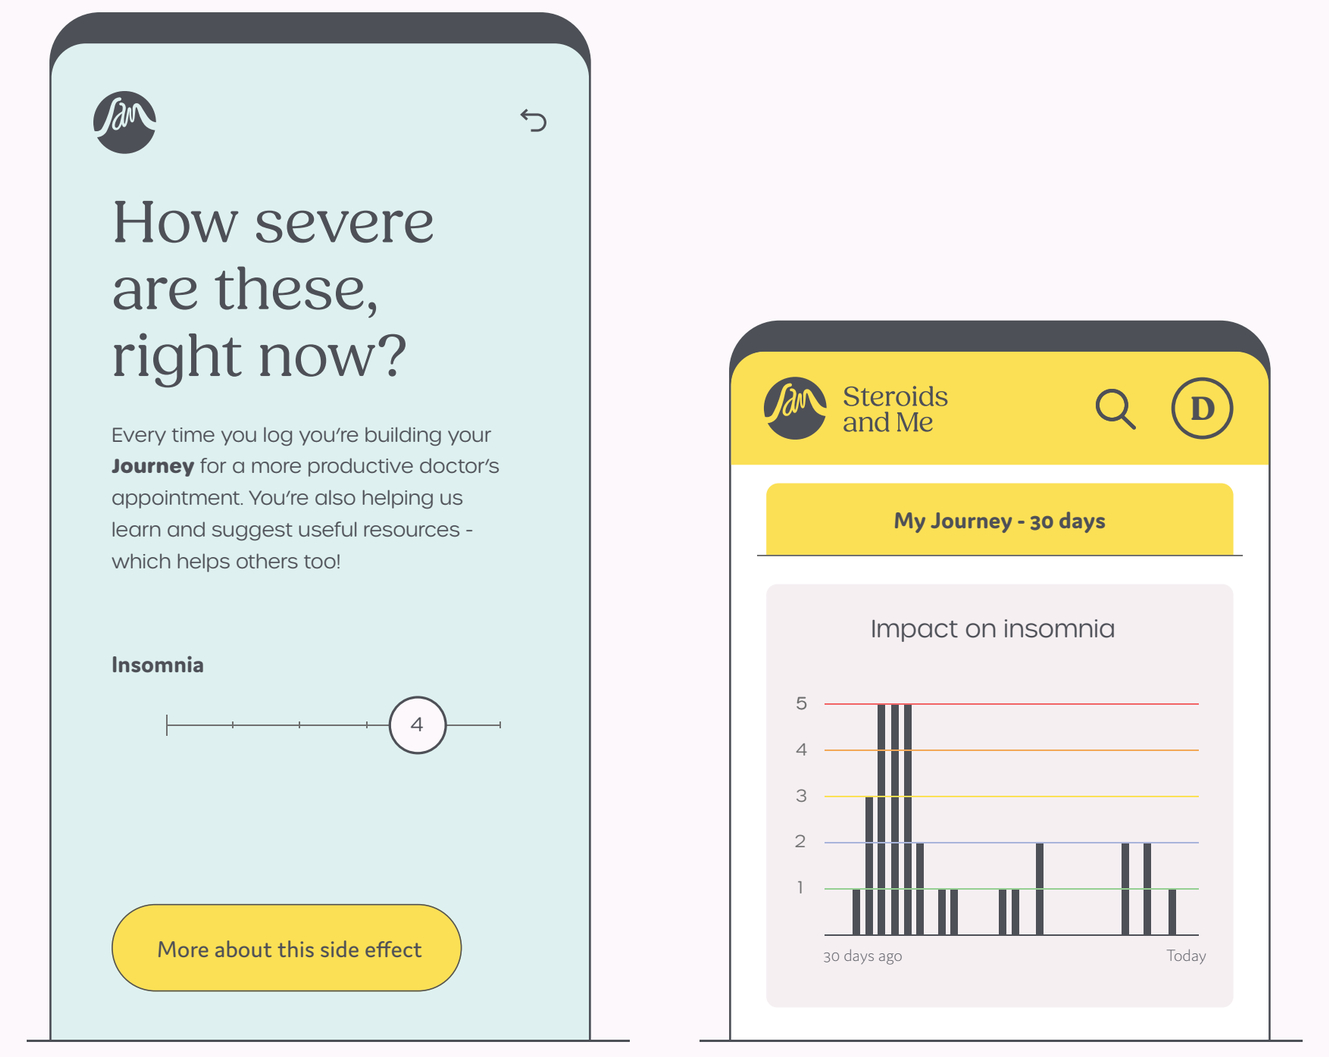

How severe are these, right now?
Steroids and Me
My Journey - 30 days
Impact on insomnia
More about this side effect

Presented at the American College of Rheumatology Annual Meeting; November 14-19 2024, Washington, DC